191: Tissue assessment of therapeutic responses to neoadjuvant SCRT with and without anti-CD40 immunotherapy sotigalimab (sotiga) in rectal cancer

ASCO Gastrointestinal Cancers Symposium

UT Southwestern Medical Center

Eslam Elghonaimy Ph.D.[1], Nina Sanford M.D.[1], Adel Kardosh M.D. Ph.D.[2], Syed A. Kazmi M.D.[3], Javier Salgado Pogacnik M.D.[4], Aaron Scott M.D. [5], Ravi Paluri M.D.[6], Sitaram Chilakamarry M.D.[4], Salwan Al Mutar M.D.[3], Amy Little Jones M.D. [3], Georgios Karagkounis M.D.[4], Fadwa Ali M.D.[4], Emina Huang M.D.[4], Brendan Paulman[1], Cassandra Hamilton[1], Xiaodong Yang M.D. Ph.D.[7], Robert Timmerman M.D.[1], and Todd A. Aguilera M.D. Ph.D.[1]

[1]Department of Radiation Oncology, [3]Department of Medicine, [4]Department of Surgery, UT Southwestern Medical Center, Dallas TX; [2]Department of Medicine, OHSU, Portland OR; [5]University of Arizona, Tuscon Az; [6]Wake Forest, Charlotte NC, [7]Apexigen, Inc., San Carlos CA

anti-CD40 immunotherapy opportunity

Background: There has been tremendous progress in the management of rectal cancer in the neoadjuvant setting and in non-operative management (NOM). However, there is a critical need to personalize systemic therapy and radiotherapy to achieve responses conducive for NOM. This study was initiated to profile immune changes to therapy in serial tumor tissue, blood, and stool from the INNATE trial (NCT04130854).

Methods: INNATE, is a multi-institutional Phase II randomized trial for patients with Stage III or high-risk Stage II rectal cancer. Patients were randomized 3:2 to receive daily short course radiotherapy (SCRT- 5 Gy x 5) and 6 cycles of FOLFOX +/- 6 infusions of sotiga, an anti-CD40 agonist, followed by surgery. The primary end point is the pathologic complete response rate and additional clinical endpoints are being assessed. Biopsies of tumor and adjacent tissue before and 2 weeks after SCRT +/- sotiga prior to chemotherapy were obtained from consenting patients. Fresh tissue was digested into single cell suspension for scRNAseq with proteomic and immune repertoire analysis. Fixed tissue was processed for histology and multiplexed immunofluorescence. Here, we report integrated and batch effect removed data from 9 patients pre- and post-therapy and analyzed patients as their own control.

Results: A total of 25 of 30 patients were enrolled at UTSW, 21 of whom underwent pre- and post-SCRT biopsies. The study population was diverse: 11 patients (37%) were Hispanic and 15 (50%) uninsured receiving care at the Dallas County hospital, Parkland. The median age was 55 (27-75) years and 13 (43.3%) were diagnosed prior to age 50. From integrated scRNAseq, we observed a significant induction of genes associated with M1- and M2-like macrophages, and dendritic cell antigen presentation in the SCRT alone group. This was accompanied by a robust Th1 and B cell response, but not T cell mediated cytotoxicity. In the SCRT + sotiga group there was a greater induction of genes associated with M1-like macrophages and dendritic cell antigen presentation, with no change in M2 genes. In lymphocytes, sotiga treatment led to an induction of genes related to Th1 and B cell response. Unlike SCRT, the combination therapy group had evidence of induced T cell mediated cytotoxicity. To better understand individual responses to therapy, we analyzed pre- and post-treatment samples for each patient. This enabled identification of features associated with poor and robust response in each treatment group.

Conclusions: On treatment biopsies during neoadjuvant therapy for rectal cancer enables deep assessment of therapeutic response, provides insight into action of investigational agents, and may identify therapy specific predictive biomarkers. Our data shows that SCRT can induce immune responses and adaptive immune responses can be further induced by sotiga.

αCD40 in the Tumor Microenvironment

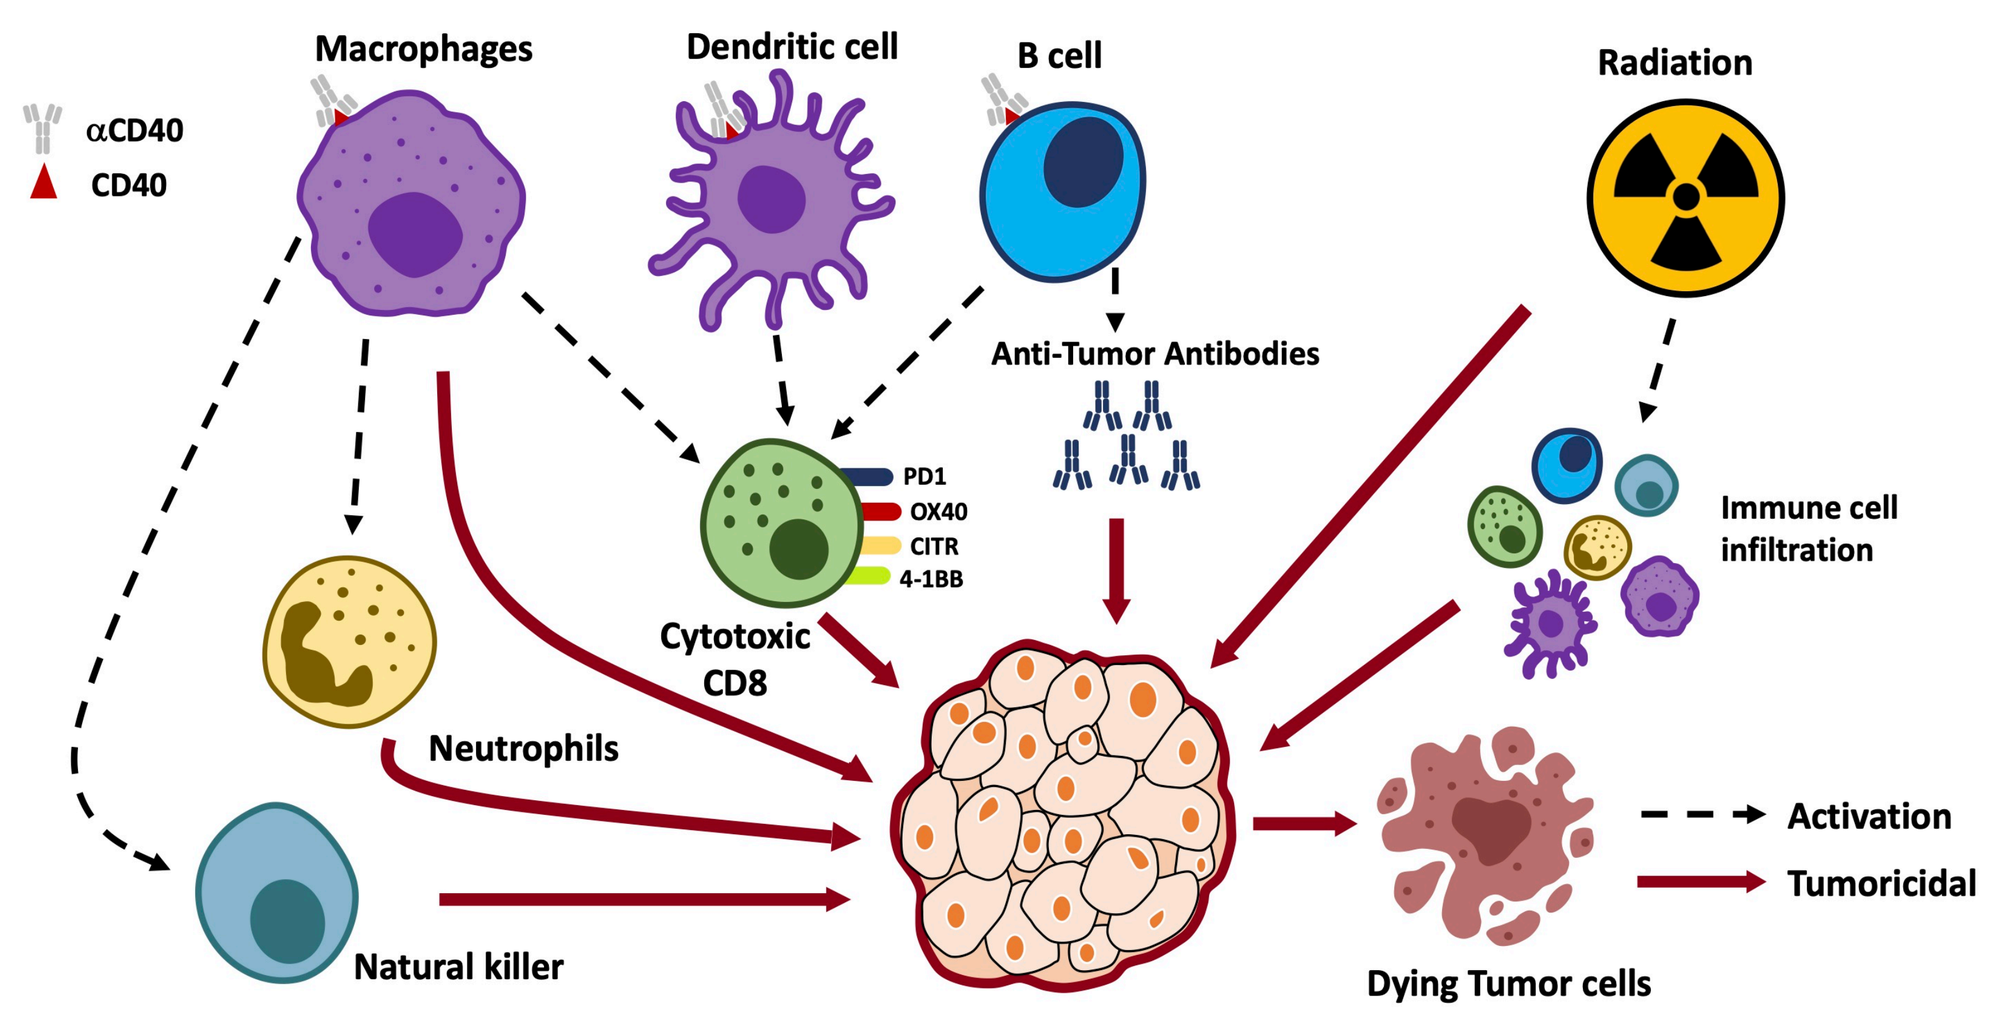

αCD40
CD40
Macrophages
Dendritic cell
B cell
Radiation
Anti-Tumor Antibodies
PD1
OX40
CITR
4-1BB
Cytotoxic CD8
Immune cell infiltration
Neutrophils
Natural killer
Activation
Tumoricidal
Dying Tumor cells

anti-CD40 can impact many players in the tumor microenvironment (TME) in combination with RT

The Drug

- Sotigalimab (sotiga): Agonistic anti-CD40 Ab with Fc mutation to enhance its potency and limit ADCC activity (*Filbert, Cancer Imm Immunother, 2021*)
- Potential activity in pancreatic cancer (*O'Hara, Lancet Oncology, 2021*)

INNATE: sotigalimab randomized in rectal cancer

Design

- Stage III or high-risk stage II (<1cm to anal ring, cT4, 3mm MR fascia, no sphincter preservation, and vascular invasion)
- Short course radiotherapy
- 3 months of FOLFOX
- sotigalimab: 3rd day post RT and chemo cycles 1-5

Endpoints

- Primary: pCR
- Secondary: 3 yr OS, 3 yr DFS

Updates

- 4 centers (UTSW, OHSU, Wake Forest, Arizona)
- 30 enrolled of *58*
- Interim pending

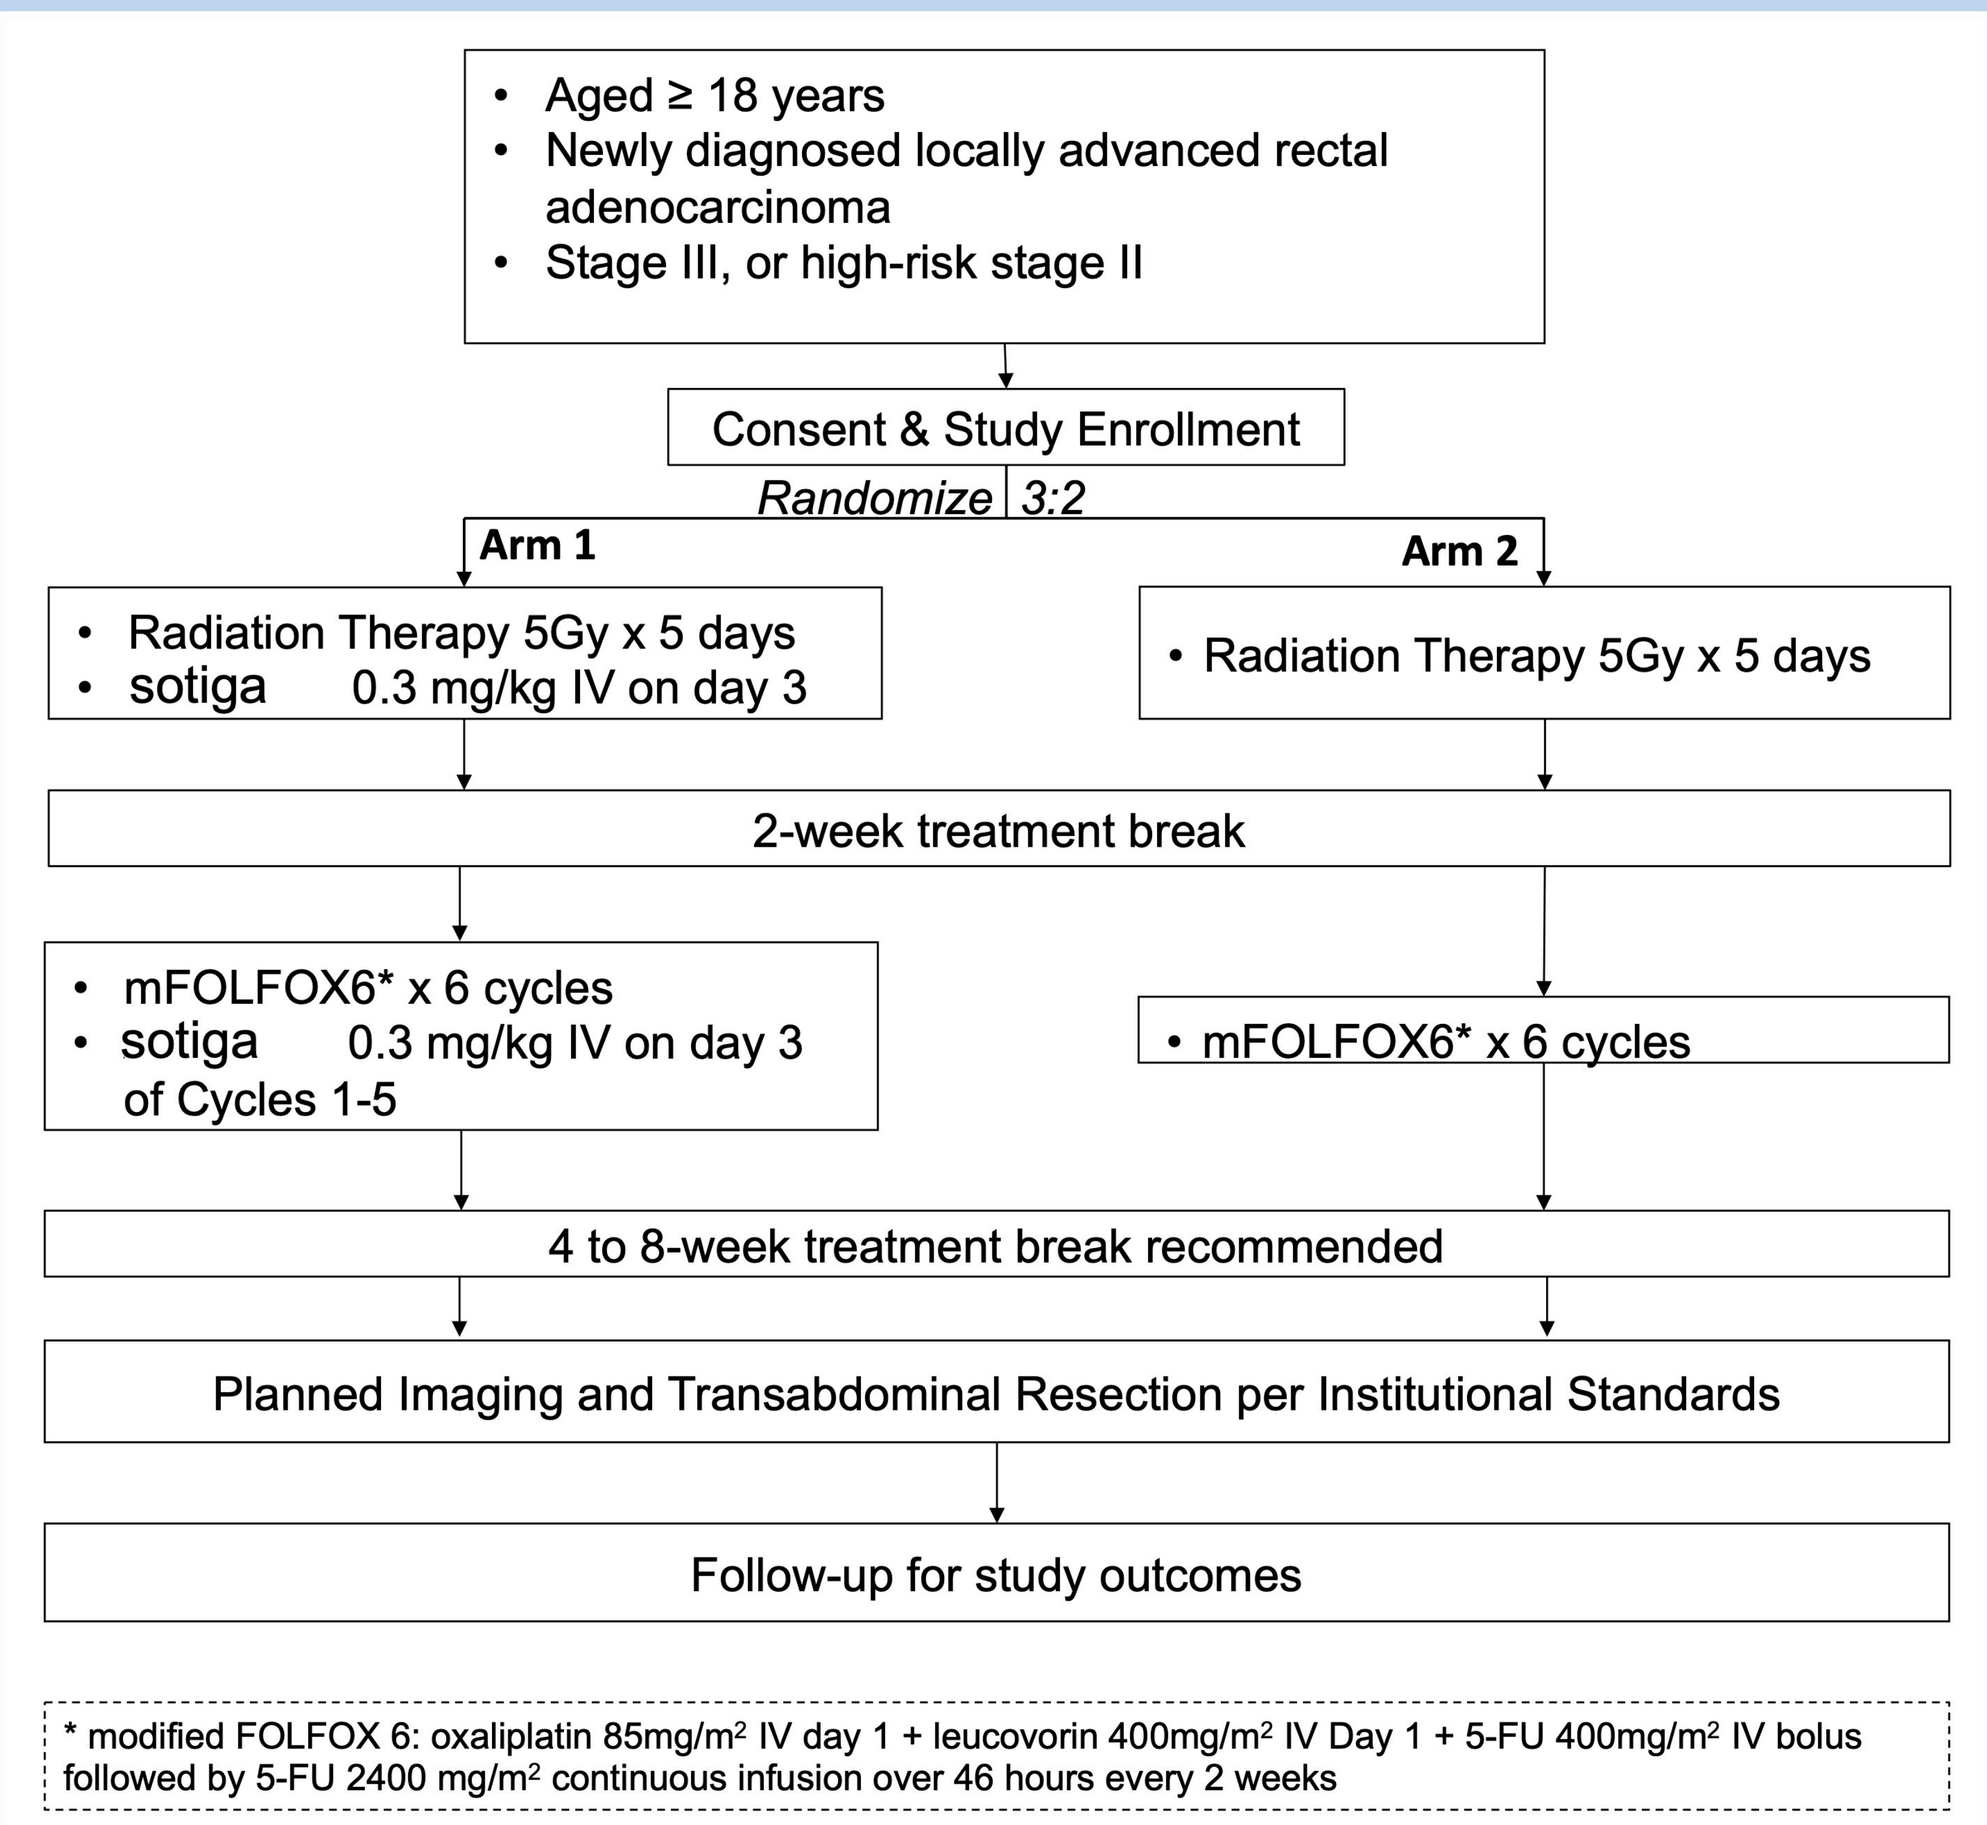

• Aged ≥ 18 years
• Newly diagnosed locally advanced rectal adenocarcinoma
• Stage III, or high-risk stage II
Consent & Study Enrollment
Randomize 3:2
Arm 1
Arm 2
• Radiation Therapy 5Gy x 5 days
• sotiga 0.3 mg/kg IV on day 3
• Radiation Therapy 5Gy x 5 days
2-week treatment break
• mFOLFOX6* x 6 cycles
• sotiga 0.3 mg/kg IV on day 3 of Cycles 1-5
• mFOLFOX6* x 6 cycles
4 to 8-week treatment break recommended
Planned Imaging and Transabdominal Resection per Institutional Standards
Follow-up for study outcomes
* modified FOLFOX 6: oxaliplatin 85mg/m² IV day 1 + leucovorin 400mg/m² IV Day 1 + 5-FU 400mg/m² IV bolus followed by 5-FU 2400 mg/m² continuous infusion over 46 hours every 2 weeks

We must innovate to improve responses in rectal cancer to avoid surgery…

- ***studying tissue responses will guide therapeutic development***
- ***sotiga induces robust immune responses***
- ***SCRT provides an immunologic opportunity***

Conclusions

- ***Sotiga + RT induces T and B cell responses greater than SCRT alone***
- ***SCRT can induce adaptive immune response in some patients***
- ***Clinical results pending***

INNATE Trial (NCT04130854) tissue collection

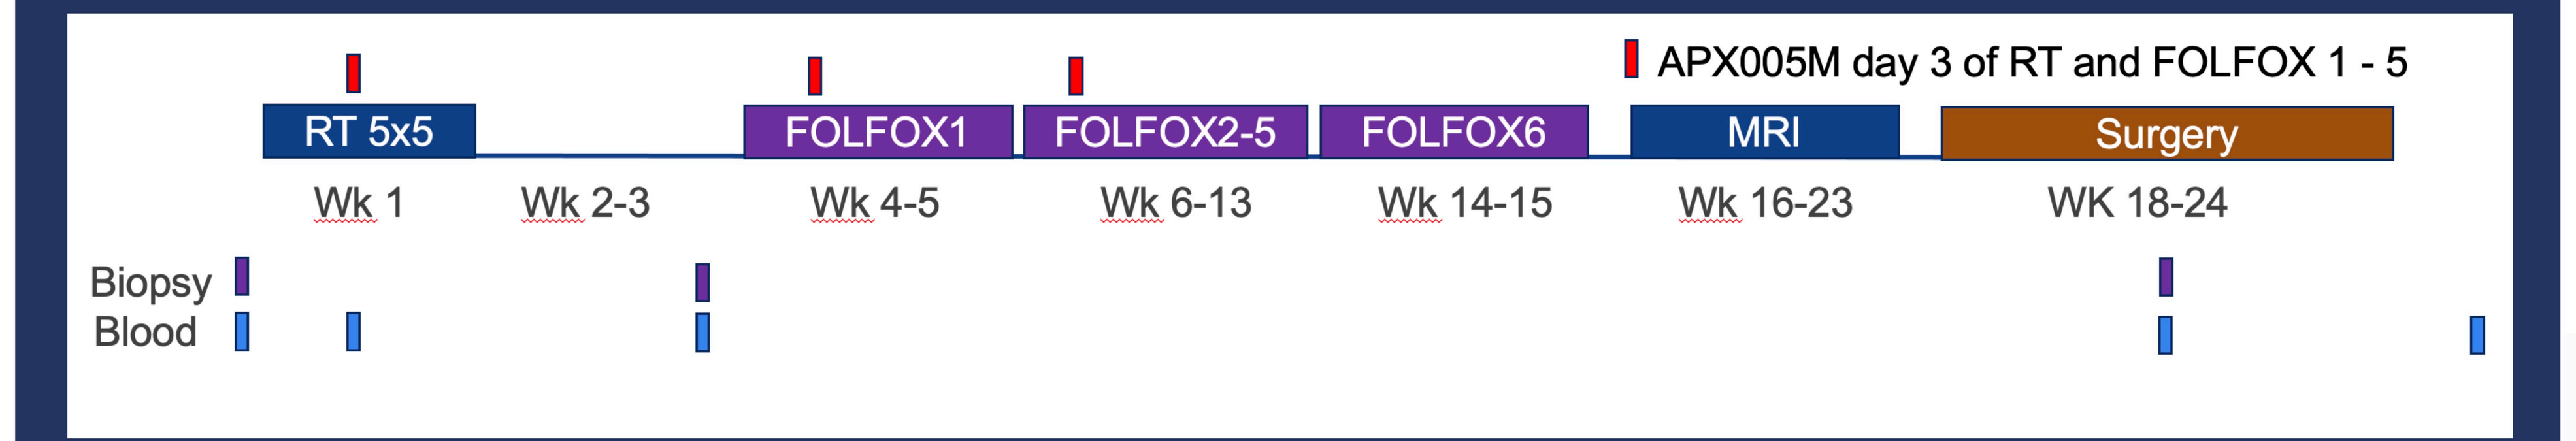

APX005M day 3 of RT and FOLFOX 1 - 5
RT 5x5
FOLFOX1
FOLFOX2-5
FOLFOX6
MRI
Surgery
Wk 1
Wk 2-3
Wk 4-5
Wk 6-13
Wk 14-15
Wk 16-23
WK 18-24
Biopsy
Blood

Enrollment to date	30 patients
Hispanic	11 (37%)
Parkland Safety Net	15 (50%)
Median age	55 (27-75) years old

Todd Aguilera: todd.aguilera@utsouthwestern.edu

Funding: Apexigen, Inc., CPRIT First-time tenure-track faculty award (TAA), and Damon Runyon Clinical Investigator Award (TAA)

Tissue analysis plan

21/25 pts at UTSW had pre/post biopsies

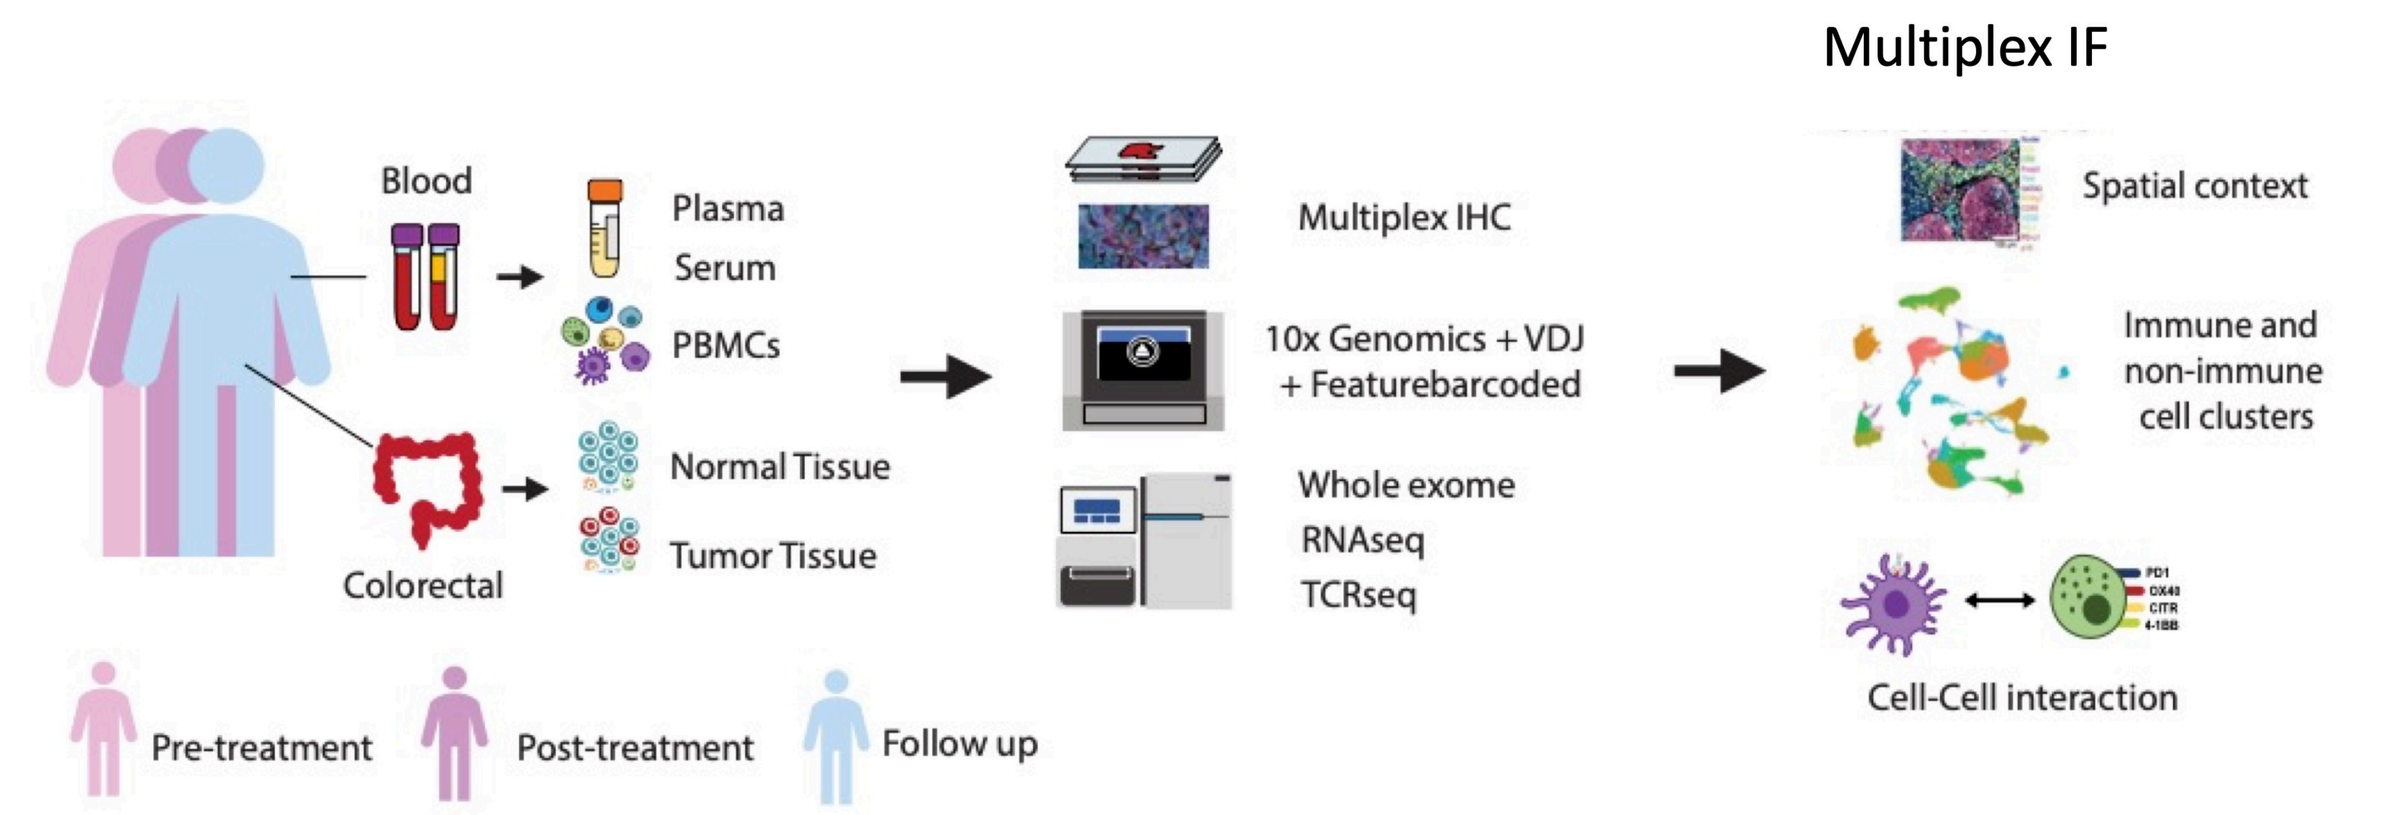

Multiplex IF
Blood
Plasma
Serum
PBMCs
Normal Tissue
Tumor Tissue
Colorectal
Multiplex IHC
10x Genomics + VDJ + Featurebarcoded
Whole exome
RNAseq
TCRseq
Spatial context
Immune and non-immune cell clusters
Cell-Cell interaction
Pre-treatment
Post-treatment
Follow up

Methods

- Biopsies (fresh, frozen, fixed)
- scRNAseq pre and post
 - TCR/BCR/CITEseq
- Bulk RNAseq and WES
- Multiplex IF pending
- Below… scRNAseq of pre and post SCRT biopsies
 - 5 Patients with RT + sotiga
 - 4 patients with RT alone

Robust dendritic cell and T cell response with sotiga

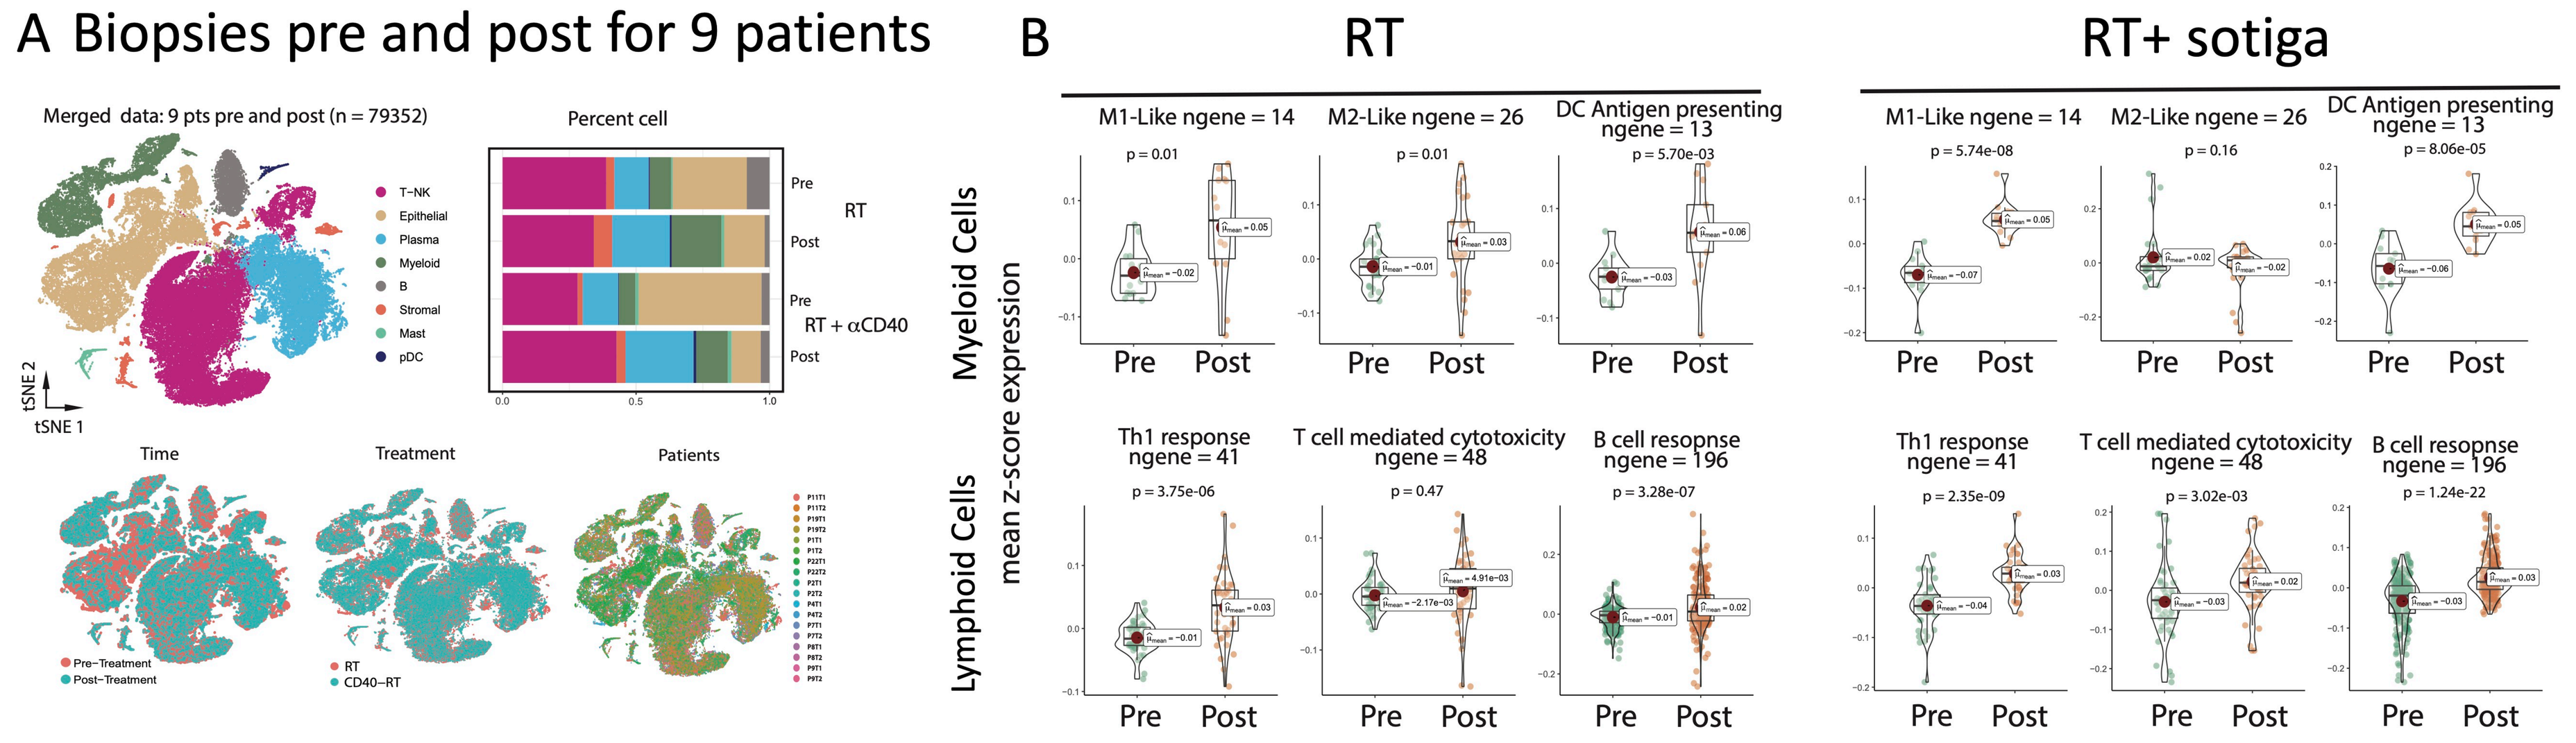

A Biopsies pre and post for 9 patients
Merged data: 9 pts pre and post (n = 79352)
Percent cell
T-NK
Epithelial
Plasma
Myeloid
B
Stromal
Mast
pDC
Pre
Post
RT
RT + αCD40
Time
Treatment
Patients
Pre-Treatment
Post-Treatment
CD40-RT
B
RT
RT+ sotiga
Myeloid Cells
Lymphoid Cells
mean z-score expression
M1-Like ngene = 14
M2-Like ngene = 26
DC Antigen presenting ngene = 13
Th1 response ngene = 41
T cell mediated cytotoxicity ngene = 48
B cell resopnse ngene = 196

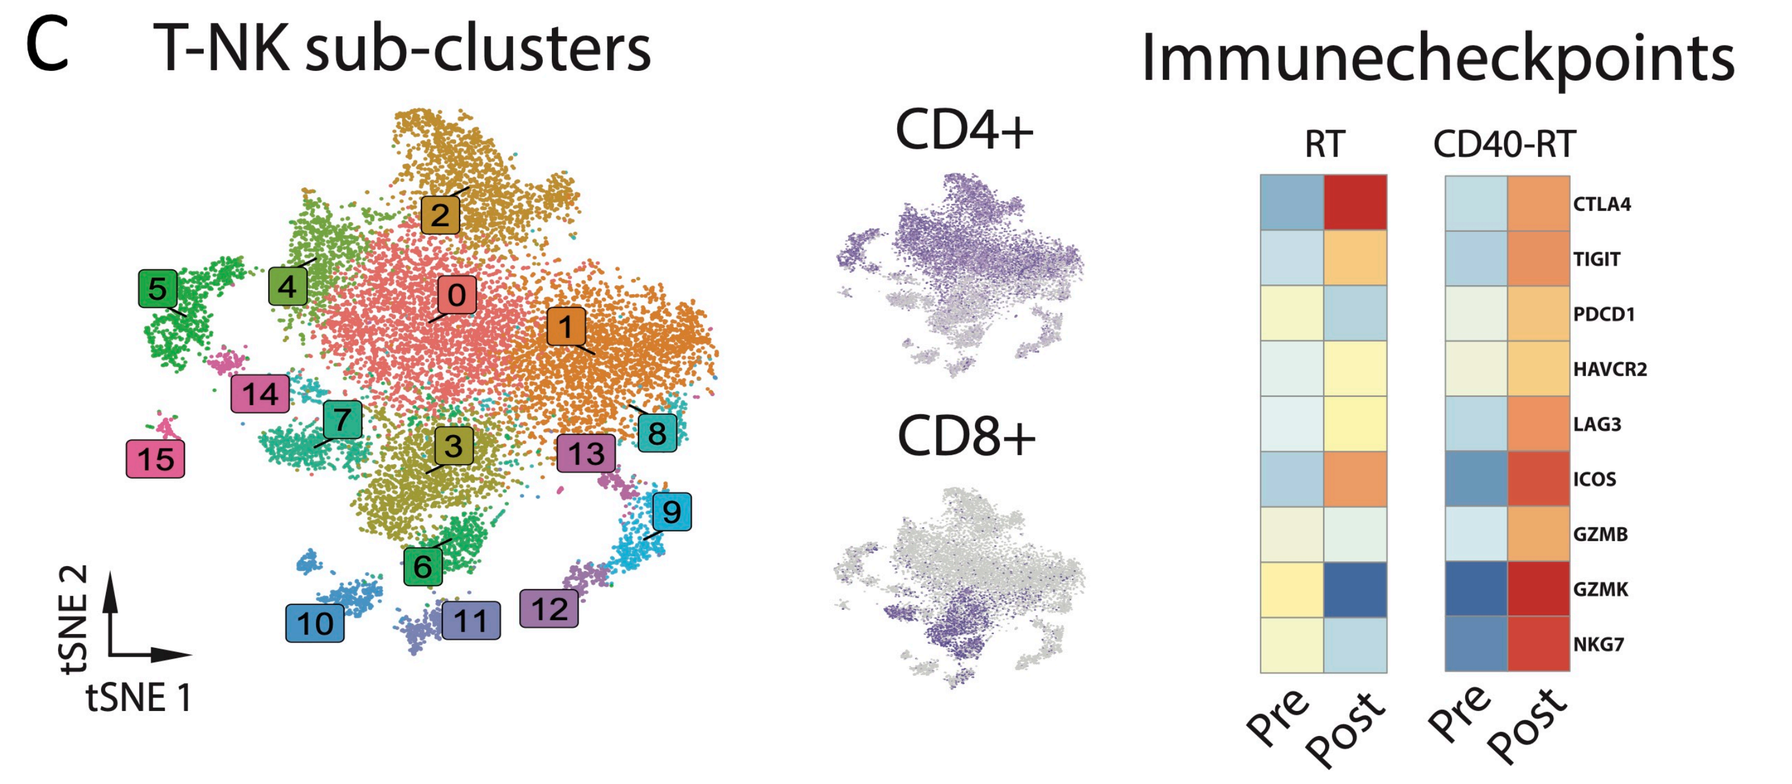

C T-NK sub-clusters
CD4+
CD8+
Immunecheckpoints
RT
CD40-RT
CTLA4
TIGIT
PDCD1
HAVCR2
LAG3
ICOS
GZMB
GZMK
NKG7
Pre
Post

A Merged data from 9 patients (5 RT+ sotiga and 4 RT alone) show equal distribution across timepoint and treatment

B Myeloid and lymphoid cell gene pathways induced with therapy in RT and RT + sotiga groups. Significant induction with RT and combo but T cell mediated cytotoxicity and B cell response greater in the combo

C T-NK cell clusters showing CD4⁺ and CD8⁺ T cells. When evaluating expression of immune checkpoints and effector genes much greater induction post sotigalimab

Adaptive immune response in a patient after RT

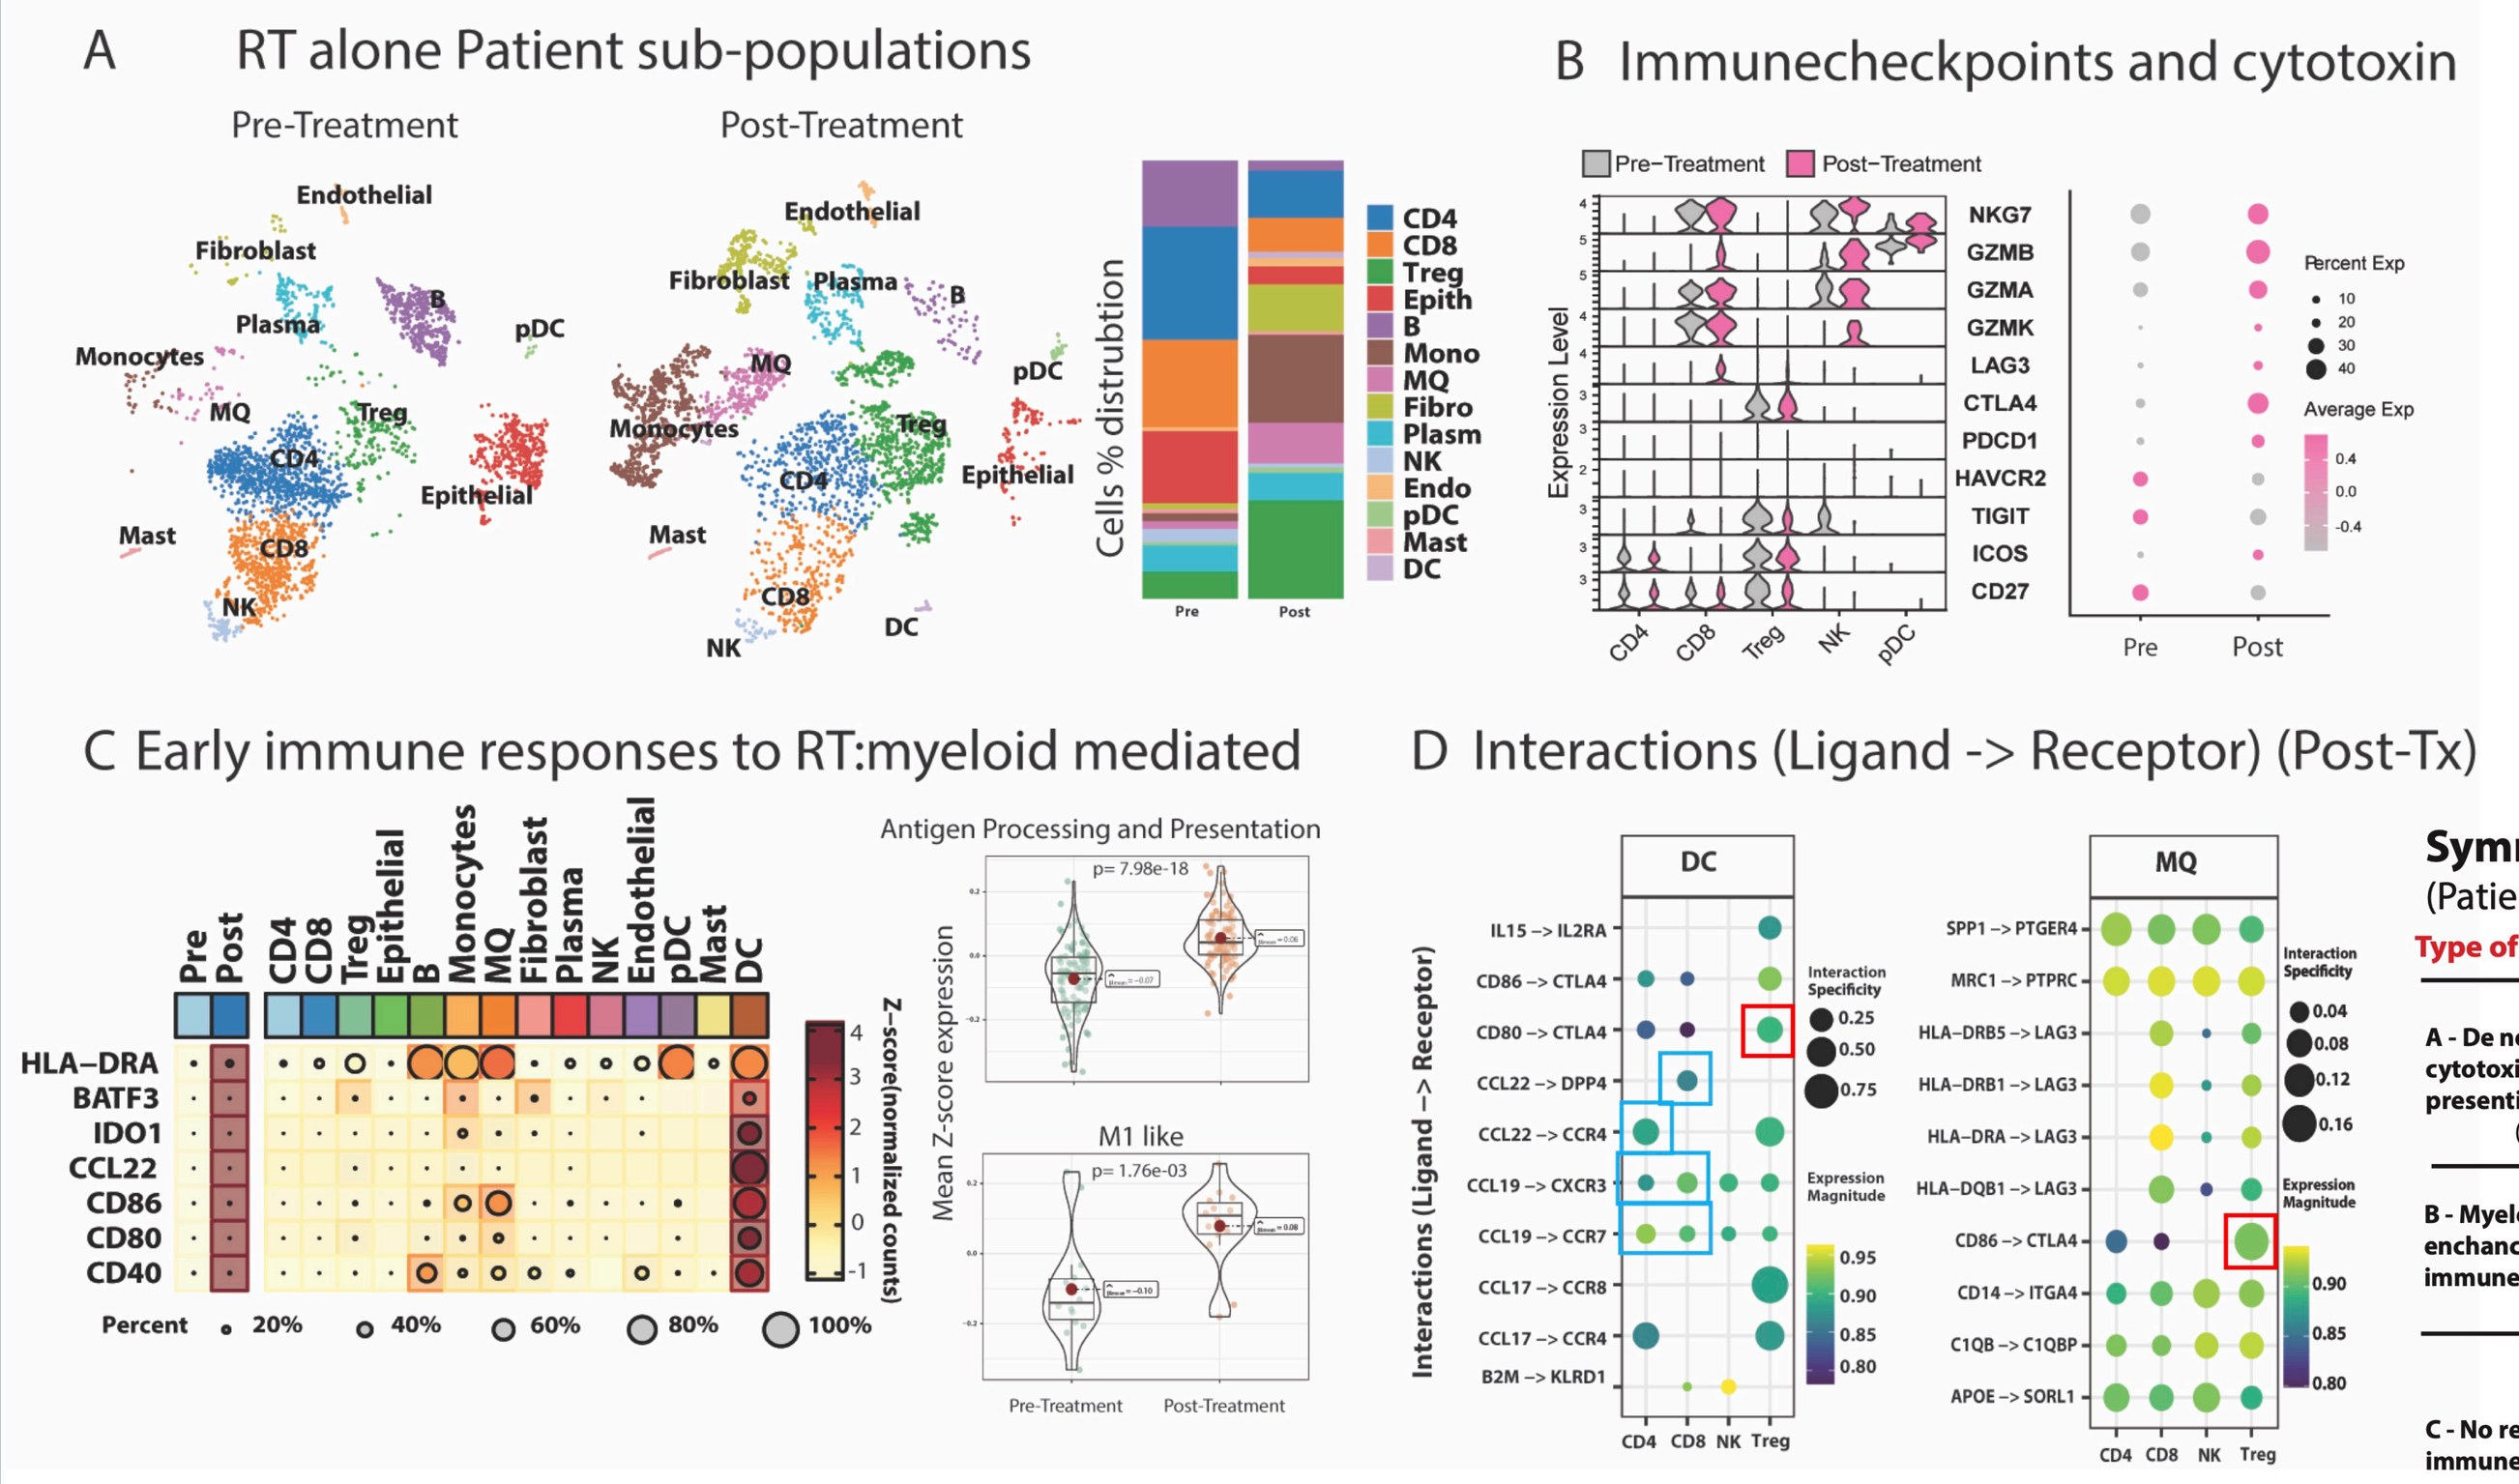

A RT alone Patient sub-populations
Pre-Treatment
Post-Treatment
Cells % distrubtion
B Immunecheckpoints and cytotoxin
Pre-Treatment
Post-Treatment
NKG7
GZMB
GZMA
GZMK
LAG3
CTLA4
PDCD1
HAVCR2
TIGIT
ICOS
CD27
C Early immune responses to RT:myeloid mediated
HLA–DRA
BATF3
IDO1
CCL22
CD86
CD80
CD40
Antigen Processing and Presentation
M1 like
D Interactions (Ligand -> Receptor) (Post-Tx)
DC
MQ

A scRNAseq cell distribution showing Influx of inflam myeloid cells after RT
B In T-NK cells: increased GZMB, GZMK and GZMA and decreased TIGIT and TIM-3 (HAVCR2)
C After RT, Induction of DC transcripts, including antigen presentation and M-1-like
D DCs and myeloid DC-like cells act on CD8 and CD4 priming, recruiting, and activation by antigen presentation pathway, chemokines, and cytokines (CCR4, CCR7, CXCR3, DPP4). On the other hand, they act on Tregs for immune suppression via CTLA4.

Symmary of RT responses from immunologic perspective
(Patients analyzed)

Type of response after RT	Celleular signals of responses	Early response events
A - De novo immune response: cytotoxic T cells and antigen presenting cells (APCs) (see figure)	↑ GZMB, GZMA, GZMK, GLYN, NKG7; ↑ Infiltration, DC1signature, Antigen presentation and priming signals; ↑ Infiltration, M1-like signature, monocytic DC like	- Infiltration of inflammatory myeloid cells; - Activate and enhance NK and CD8 cytotoxic secretion; - Induced CD8 priming by APCs
B - Myeloid population that enchances and maintains immune response	→ GZMB, GZMA, GZMK, GLYN, NKG7; ↑ TIM-3, CTLA4, LAG3; → DC1signature, APC signaling	- Maintain cytotoxic secretion from pre-existing CD8 and NK cells by interaction with pre-existing DC
C - No response mediated by immune cells was detected	↓ GZMB, GZMA, GZMK, GLYN, NKG7; ↑ TIM-3, CTLA4, LAG3; ↑ pre-existing activation; ↓ pre-existing or infiltrated	- RT did not change the pre-existing low level of cytotoxic T cells; - No DC or Macrophages infiltration was detected after RT